BNC210: A Novel Compound with Potent Anxiolytic Activity

NEUROFIT PRECLINICAL RESEARCH

E. Andriambeloson[1], S. Wagner[1], B. Huyard[1], B. Sleebs[2], N. Quasi[2], C. Bui[2], S. O'Connor[2] and I. Street[2]

[1]Neurofit SAS, Illkirch, France; [2]Bionomics Ltd, Adelaide, Australia

Introduction

Anxiety is the most widespread of all mental disorders, and current treatments are still limited by presence of clinically undesirable side-effects of anxiolytic drugs . So, the discovery of novel, fast-acting anxiolytic, which is without significant side-effects (particularly sedation), would be welcomed. By applying a targeted medicinal chemistry strategy beginning from a compound cited in the literature, Bionomics has developed BNC210. This novel compound has potent anxiolytic activity (0.1mg/kg) in a range of rodent models and has no effect on short term memory at doses up to 20mg/kg or on spontaneous motor activity at doses up to 100 mg/kg thus realising a therapeutic index of over 1,000. BNC210 is readily absorbed and has good bioavailability in rats (69%) with a half life of 6.2 h. The molecular mechanism of BNC210 has not been fully elucidated. Early safety assessment indicates that BNC210 will be safe and well tolerated.

Methods

- BNC210 was developed in a targeted medicinal chemistry program beginning from a compound cited in the literature with newly described anxiolytic activity.
- The aim of the program was to produce compounds with improved solubility, metabolic stability, and efficacy.
- A small focused library of 42 compounds was synthesized and evaluated according to the scheme in the adjacent figure.

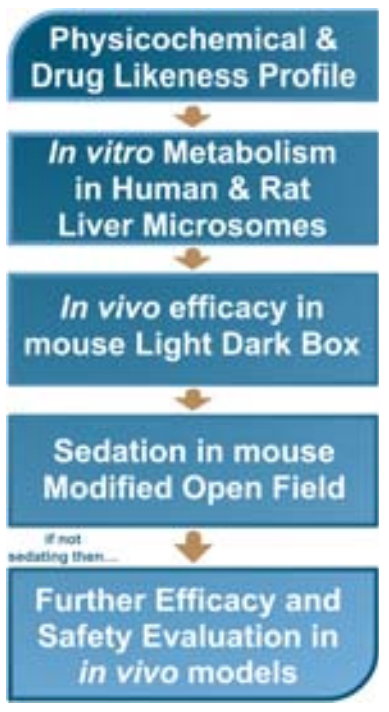
Physicochemical & Drug Likeness Profile
In vitro Metabolism in Human & Rat Liver Microsomes
In vivo efficacy in mouse Light Dark Box
Sedation in mouse Modified Open Field
if not sedating then...
Further Efficacy and Safety Evaluation in in vivo models

Physicochemical and Pharmacokinetic Properties

Early ADME-T Studies are encouraging:

- Good Plasma stability : PK (rat)
 - ⇒ $T_{1/2}$ = 6.2 h
 - ⇒ T_{max} = 60 min
 - ⇒ %F = 69%
- Plasma Protein Binding
 - ⇒ 77.42% Bound
- HERG – no binding at 10 μM
- No Genotoxicity (micronucleus)
- Microsome Metabolism
 - ⇒ Low to intermediate metabolism rates
- No significant species difference
- No inhibition of 5 CYP450 isoforms
 - ⇒ EC50 =>30uM

BNC210 has physicochemical properties that are consistent with good "drug like" profile:

- MW: = 418
- PSA: 75 Å^2
- FRB: 4
- logD pH7.4 = 2.71
- Solubility: pH 2.0 > 100 mg/mL
 pH 7.4 > 100 mg/mL
- Number of steps in synthesis: 8
- Number of Chiral Centres: 0

BNC210 was profiled against 106 trans-membrane and soluble receptors, ion channels and mono-amine transporters plus 36 enzymes in order to identify its molecular target(s). Significant inhibition of binding was seen at three targets.

- **NET** (Ki = 6.3 μM)
- **NK2 receptor** (Ki = 9.4 μM)
- **Adenosine A3** (Ki = 13 μM)

Two of these proteins, NK2 receptor and NET (Noradrenaline Transporter) are the targets of several antidepressant and anxiolytic compounds which are on the market or in development.

The IC50's of BNC210 at these targets do not correlate with its in vivo potency or with its pharmacology profile. Further work is in progress to define the M.O.A.

Also tested:

- CRFR1
- MCHR1
- BZD (central) – diazepam
- Chloride ion channel – picrotoxin
- GABA binding site – muscimol
- $GABA_A$ transporter – nipecotic acid
- $GABA_B$(1b) receptor – CGP

BNC210 has a Unique Pharmacology

- Potent anxiolytic
- No sedation or ataxia
- No development of tolerance
- Not addictive
- No memory impairment

BNC210 is a novel proprietary compound

- **Belongs to a chemical class which has a long history of safe pharmaceutical use**
- **Early safety assessment indicates that BNC210 is safe and will be well tolerated**

Results

BNC210 demonstrates a potent anxiolytic activity in 3 anxiety tests:

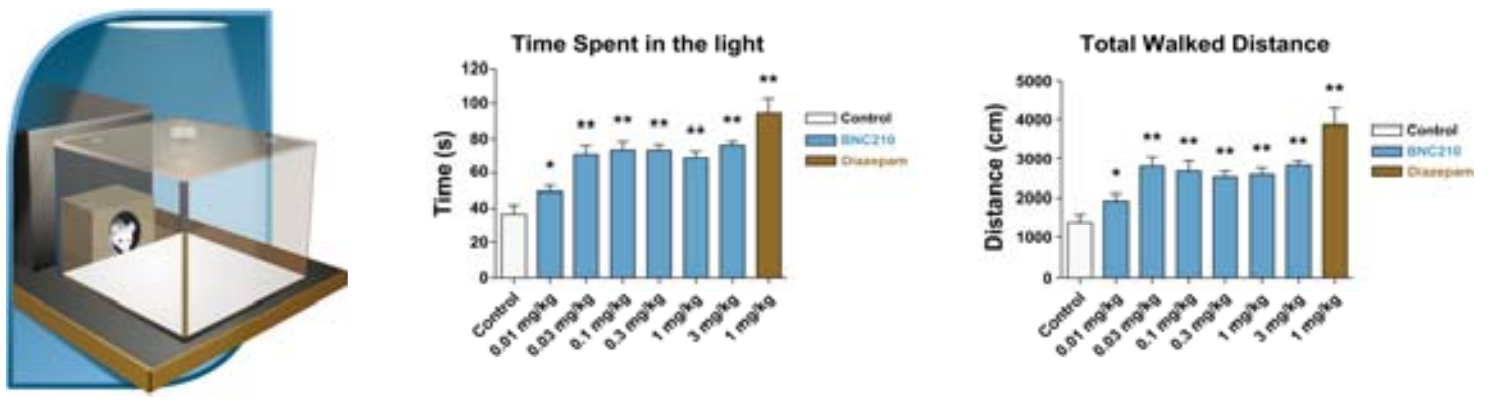
Time Spent in the light
Time (s)
Total Walked Distance
Distance (cm)
Control
BNC210
Diazepam
Control
0.01 mg/kg
0.03 mg/kg
0.1 mg/kg
0.3 mg/kg
1 mg/kg
3 mg/kg
1 mg/kg

BNC210 gives an outstanding performance in the Light Dark Box. At doses as low as 0.01mg/kg, it significantly increases the amount of time that mice spend in, and the distance walked in, the brightly lit chamber.
Data represent mean ± SEM. n=10 mice, *p=<0.05, **p<0.01 (Fishers Protected Least Significant Difference test).

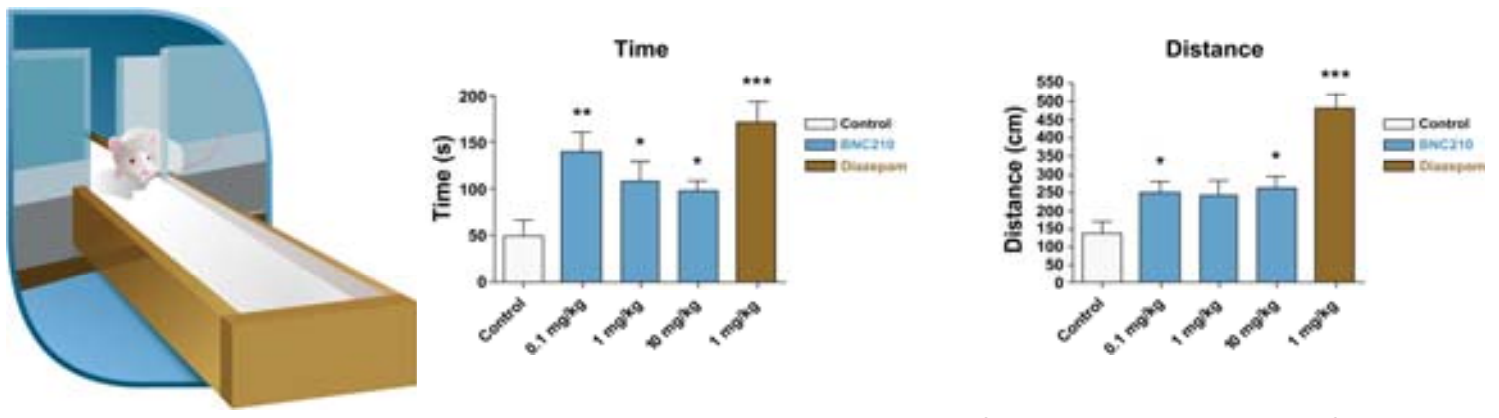
Time
Time (s)
Distance
Distance (cm)
Control
BNC210
Diazepam
Control
0.1 mg/kg
1 mg/kg
10 mg/kg
1 mg/kg

BNC210 has a potent anxiolytic effect when examined in the Rat Elevated Plus Maze. Compared to control animals, BNC210 increased the time spent and the distance travelled in the open arms of the maze. The minimum effective dose in this model was 0.1mg/ kg.
Data represent mean ± SEM. n=10 rats, *p<0.05, **p<0.01, ***p<0.001 (Fishers Protected Least Significant Difference test).

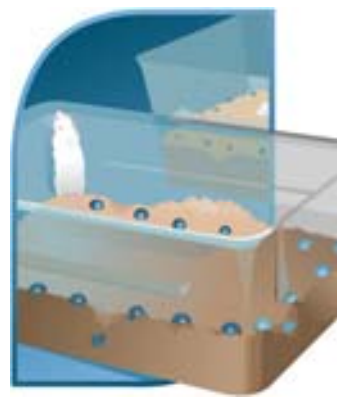

The **Marble Burying Test** is used as a model for both anxiety and obsessive compulsive disorders. Mice have a natural tendency to bury marbles under their bedding when placed in a cage with rows of evenly spaced marbles on the floor. Suppression of spontaneous burying has been used as a measure of the anxiolytic action of drugs. Mice treated with benzodiazepines and classes of antidepressants, bury less marbles when compared to the control mice

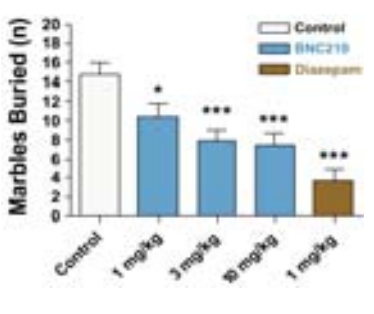
Marbles Buried (n)
Control
BNC210
Diazepam
Control
1 mg/kg
3 mg/kg
10 mg/kg
1 mg/kg

BNC210 has a significant effect on Marble Burying behaviour in mice, reducing their innate tendency for marble burying by more than 50% at a dose of 10mg/kg and a clear dose response was seen in the dose range used in the experiment.
Data represent mean ± SEM. n=10 mice, *p=<0.05, ***p<0.001 (Fishers Protected Least Significant Difference test).

No side effects

BNC210 is not sedating in **mice** in the **Open-Field**

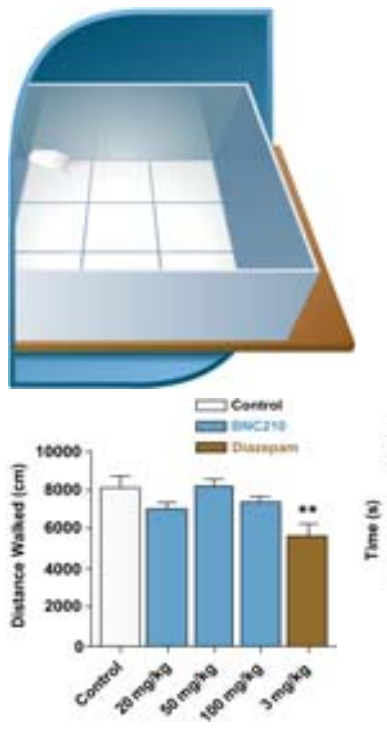
Distance Walked (cm)
Control
BNC210
Diazepam
Control
20 mg/kg
50 mg/kg
100 mg/kg
3 mg/kg

BNC210 does not show any sign of sedation in the Open-Field (dark) at doses of 20, 50 and 100mg/kg as evidenced by the lack of effect on spontaneous locomotor activity. The sedative side effect of diazepam is clearly observed at a dose of 3mg/kg. Data represent mean ± SEM. n=10 mice, **p=<0.01 (Fishers Protected Least Significant Difference test).

BNC210 does not affect motor coordination in **mice** at high doses in the **Rotarod Test**

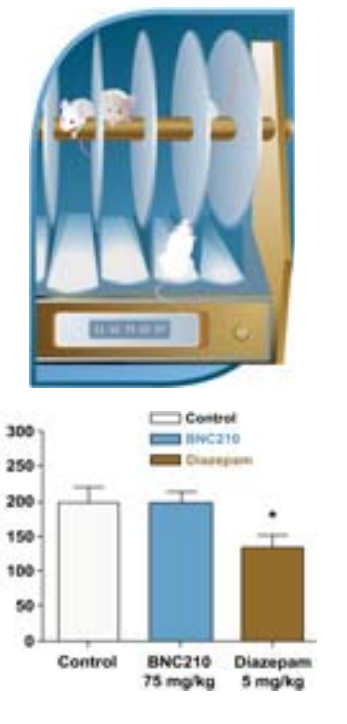
Time (s)
Control
BNC210
Diazepam
Control
BNC210 75 mg/kg
Diazepam 5 mg/kg

There is no difference in the length of time that control mice and mice treated with 75mg/kg of BNC210, stay on the Rotarod. In contrast, 5mg/kg of diazepam reduces the ability of mice to stay on the rotating rod by almost 50% (n=10 mice).
Data represent mean ± SEM. n=10 mice, *p=<0.05 (Fishers Protected Least Significant Difference test). Experiment performed by Dr P. Davies, Howard Florey Institute

BNC210 is not addictive in **mice** in the **Conditioned Place Preference Test**

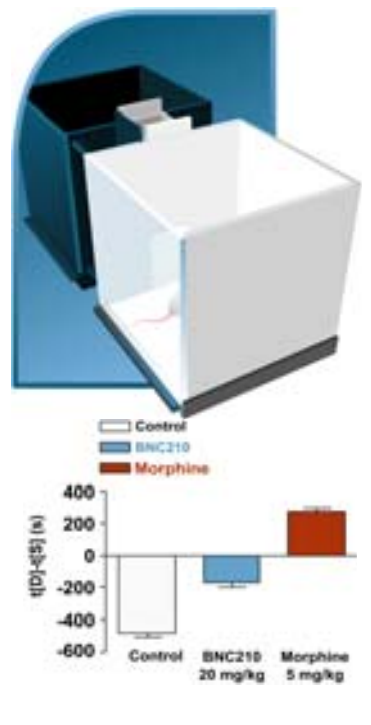
Control
BNC210
Morphine
t[D]-t[S] (s)
Control
BNC210 20 mg/kg
Morphine 5 mg/kg

BNC210 does not have addictive properties and does not increase the time mice spend in the drug-paired compartment (t[D]) of the **Conditioned Place Preference Test** when allowed to freely explore an apparatus containing a saline-paired compartment (t[S]) and a drug-paired compartment. In contrast, 5mg/kg of morphine increases the time mice spend in the drug-paired compartment by almost 40%.
Data represent mean ± SEM. n=10 mice.

BNC210 does not impair memory in **rats** in the **Object Recognition Test**

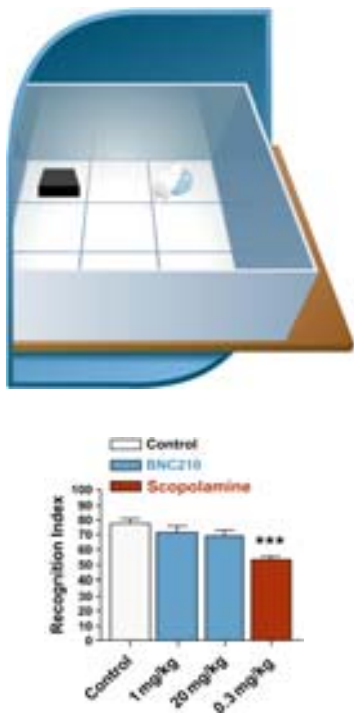
Control
BNC210
Scopolamine
Recognition Index
Control
1 mg/kg
20 mg/kg
0.3 mg/kg

BNC210 does induce memory impairement in rats in the **Object Recognition Test** at doses 200-2000 times larger than those required for its anxiolytic effect.
Data represent mean ± SEM. n=10 rats, ***p<0.001 (Fishers Protected Least Significant Difference test).

Chronic Administration (14-days) of BNC210 does not cause tolerance to its anxiolytic activity in MICE.

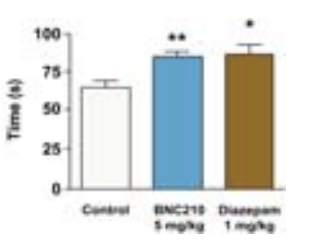
Time (s)
Control
BNC210 5 mg/kg
Diazepam 1 mg/kg

Male Swiss Mice were dosed daily by oral gavage with 5mg/kg of BNC210 for a period of 14 days. The mice were then evaluated in the **Light Dark Model** of anxiety. BNC210 had a clear anxiolytic effect and there was **no evidence for the development of tolerance to the drug after chronic treatment**.
Data represents the mean ± SEM of 10 mice and is analysed by Fisher's Protected Least Significant Difference test. *p≤0.05, **p≤0.01

Chronic Administration (14-days) of BNC210 does not cause sedation in MICE.

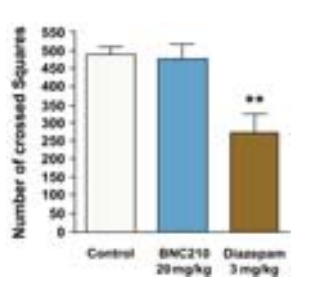
Number of crossed Squares
Control
BNC210 20 mg/kg
Diazepam 3 mg/kg

Male Swiss Mice were dosed daily by oral gavage with 20mg/kg of BNC210 for a period of 14 days. The mice were then evaluated in the **Open-Field (DARK)** for evidence of sedation. **Mice did not show any sedation after chronic administration** with BNC210. Data represents the mean ± SEM of 10 mice and is analysed by Fisher's Protected Least Significant Difference test. **p≤0.01

- **BNC210 has potent activity in *three rodent models of anxiety* at 0.1mg/kg**
- **Shows no memory impairment in rats - Is not sedating in mice or rats up to 100mg/kg**
- **Does not affect motor coordination in mice - Is not addictive in mice**
- **Has good physicochemical & PK properties**
- ***Current therapeutic window is >1000 in mice and rats***